ANALYSIS OF PRODUCT LABELING CHANGES FOR SUCCESSFUL PRESCRIPTION TO OVER-THE-COUNTER SWITCHES IN THE US.

Kim Le, Pharm.D.[1,2], Meena Ramachandra, Pharm.D.[1,2], Lindsay Brust, Pharm.D., BCPS[1], Vrunda Patel, Pharm.D.[2], Michael Toscani, Pharm.D.[1]

[1] Rutgers, the State University of New Jersey; [2] Bayer HealthCare, Consumer Care. Correspondence to: kim.le@bayer.com. SelfCare2014;5(5):115 - poster1

BACKGROUND

The landscape for prescription (Rx) to over-the-counter (OTC) switches has gained momentum in the past decade and is likely to continue. Currently, there is no definitive pattern of characteristics that would predict a successful Rx to OTC switch. Due to the changing Rx to OTC switch landscape, there are many potential switch candidates in a variety of therapeutic areas. A close appraisal of product labeling prior to and following a Rx to OTC switch could help detect 'switchable' characteristics and ultimately identify products with a potential for a successful switch. Although a number of published reports discuss the regulatory perspective of the switch process, the impact of switches on patient access, or the future of potential Rx to OTC switches, there were no studies found that focused specifically on the labeling in Rx to OTC switches.

OBJECTIVE

To identify labeling changes for medications that have had marketing status changes from prescription to over-the-counter, thereby characterizing successful switches

METHODS

Identified first-in-class switches based on FDA's published Prescription to Over-the-Counter (OTC) Switch List [1]

- Labels from the approximate time of switch approval were either downloaded from the FDA database of approved drug products[2] or obtained from the sponsor company.
- Unable to obtain Monistat 3 combo pack labeling, assessed Lotrimin Ultra instead.

Developed standardized questions to assess each switched product in key areas of interest based on pertinent sections in the Rx and OTC labeling.

- Areas of interest: Indications/Uses, Dosage & Administration/Directions for Use, Warnings & Precautions/Warnings, Use in Special Populations/Warnings, Miscellaneous.

Categorized questions into primary and secondary assessments.

- Primary questions broadly assessed labeling changes using binary response options, which were:

 "Not applicable": Rx labeling does not address the question at hand.

 "Preserved": Rx labeling information communicated in OTC labeling.

 "Not preserved": Rx labeling information is not communicated in OTC labeling.

- Secondary questions further quantified labeling differences identified by primary questions.

Analysis

- Utilized descriptive statistics (including frequencies, percentages, and mean scores) to analyze labeling elements of successful Rx to OTC switches by switch product and area of interest.

IRB review for either exemption or full approval was not required.

RESULTS

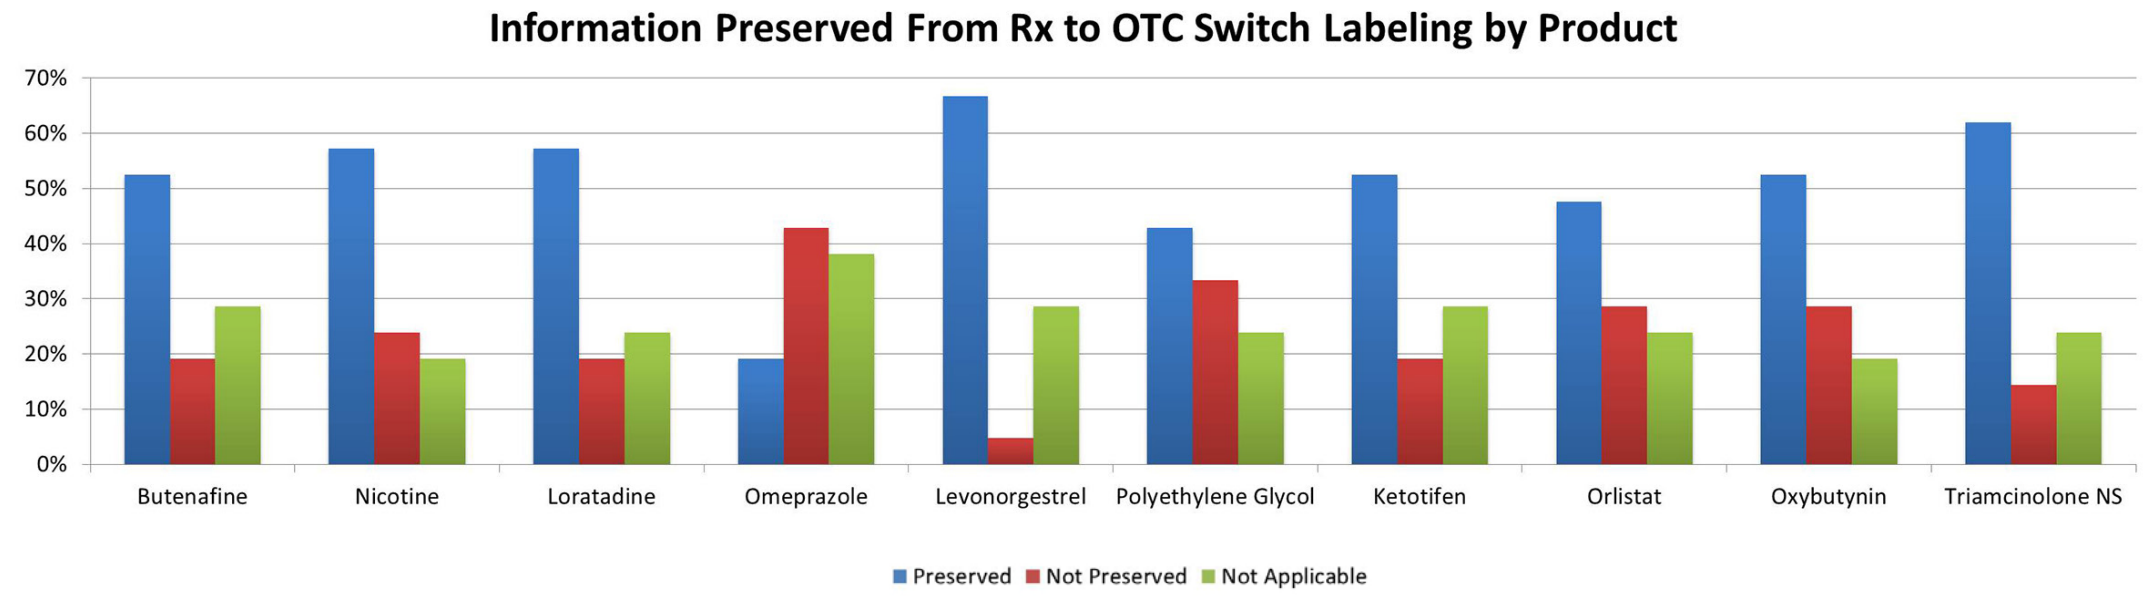
Information Preserved From Rx to OTC Switch Labeling by Product
70%
60%
50%
40%
30%
20%
10%
0%
Butenafine
Nicotine
Loratadine
Omeprazole
Levonorgestrel
Polyethylene Glycol
Ketotifen
Orlistat
Oxybutynin
Triamcinolone NS
Preserved
Not Preserved
Not Applicable

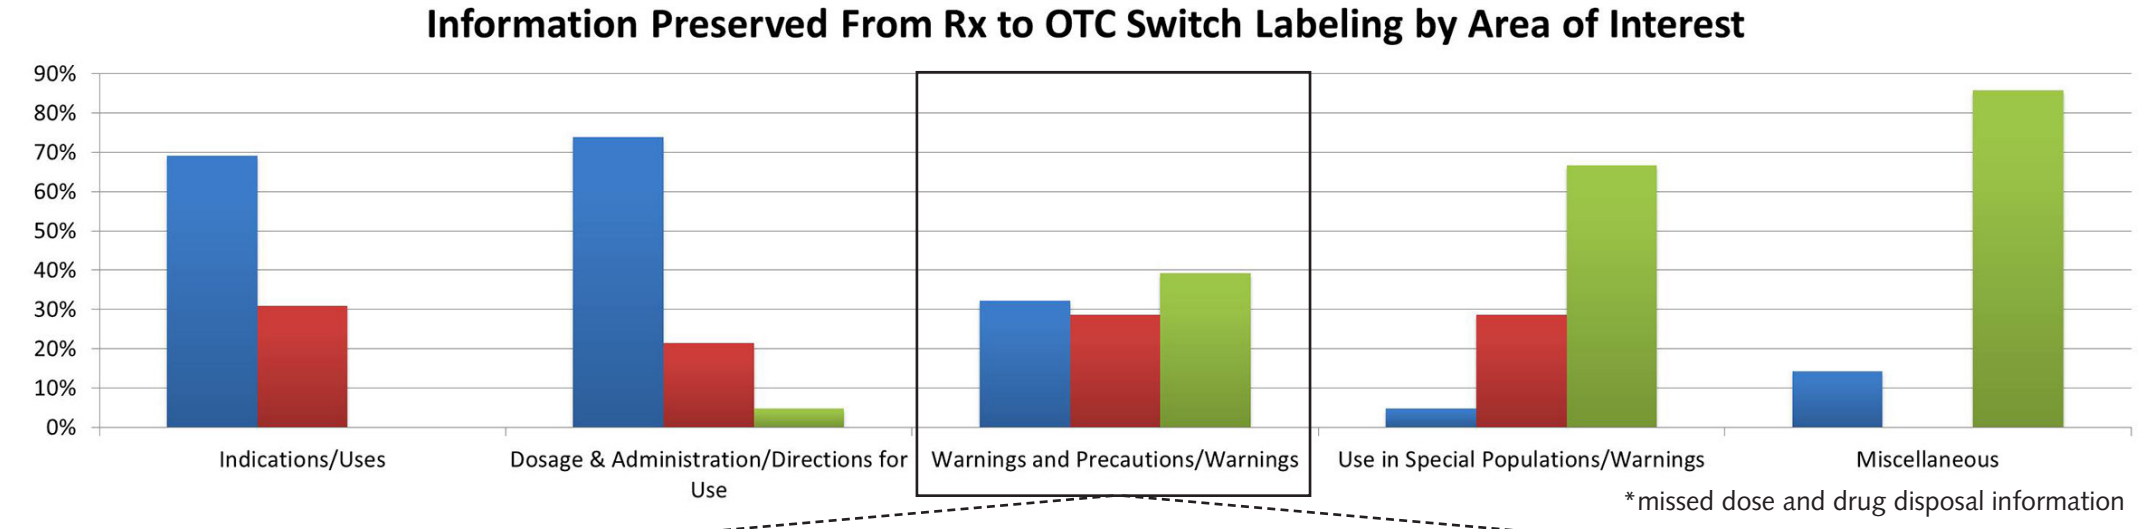
Information Preserved From Rx to OTC Switch Labeling by Area of Interest
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Indications/Uses
Dosage & Administration/Directions for Use
Warnings and Precautions/Warnings
Use in Special Populations/Warnings
Miscellaneous
*missed dose and drug disposal information

A Closer Look at Warnings and Precautions/Warnings
Information From Rx Labeling Captured in OTC Labeling

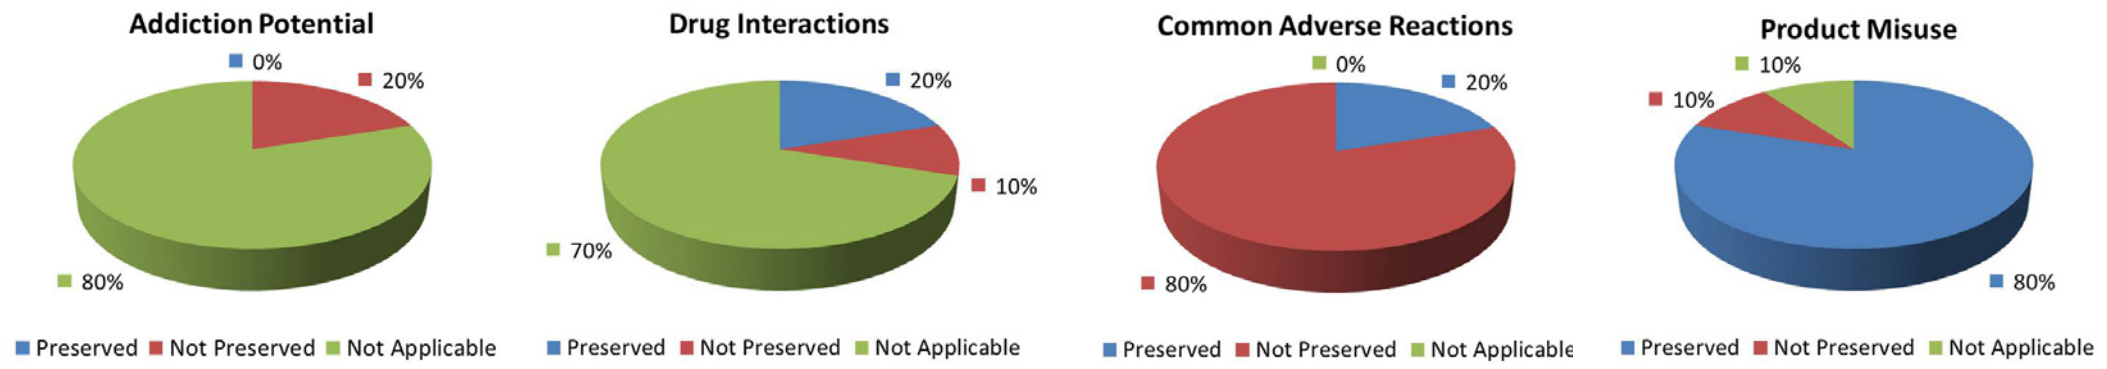
Addiction Potential
0%
20%
80%
Preserved
Not Preserved
Not Applicable
Drug Interactions
20%
10%
70%
Preserved
Not Preserved
Not Applicable
Common Adverse Reactions
0%
20%
80%
Preserved
Not Preserved
Not Applicable
Product Misuse
10%
10%
80%
Preserved
Not Preserved
Not Applicable

DISCUSSION AND LIMITATIONS

The data suggests that information from the Rx labeling is largely preserved in the OTC labeling during the switch process.

A high percentage (>50% of responses) were "Not Applicable" meaning the Rx labeling does not have specific information relevant to the following labeling elements and therefore did not need to be included in OTC labeling:

- Warnings and Precautions/Warnings: addiction potential and drug interactions.
- Use in Special Populations/Warnings: hepatic and renal dosing considerations.
- Miscellaneous: missed dose and drug disposal information

Areas of the labeling where majority of the information was transferred to the OTC labeling.

- 69% of Indications/Uses are the same as the Rx products
- 74% of Dosing & Administration/Directions for Use are the same as the Rx products.

SelfCare
Advancing the *study&understanding* of self-care

DISCUSSION AND LIMITATIONS *continued*

High percentage of common adverse events observed as "Not Preserved"

- Common adverse reactions identified in the Rx labeling are drawn from clinical trial data but may not reflect clinical relevance, which is considered in development of OTC labeling

Notable findings based on analysis by switch product

- Omeprazole has the highest percentage of "Not Preserved" because the OTC indication is not an Rx indication
- Orlistat contains a patient package insert, relatively unique to OTC products, which may affect and potentially minimize the percentage "Not Preserved"

Recurring characteristics of switched OTC product labeling (observed for ≥50% of products)

- All Rx indications not included
- Most products switched for adolescent and adult use
- Defined duration of use
- Common adverse reactions not transferred to OTC labeling
- Standardized pregnancy and lactation language employed

LIMITATIONS

- Did not evaluate unsuccessful switches due to limited public information
- Analysis required subjective interpretation of translation from healthcare professional to consumer language
- Comparison is limited to the labeling and is quantitative in nature

CONCLUSIONS

Overall the OTC labeling substantially communicates information from the Rx labeling.

"Not Applicable" responses indicate Rx labeling for switched products report minimal warnings regarding addiction potential, drug interactions, use in special populations, missed doses, and drug disposal instructions.

Variability in results suggests there is no standard to define what Rx labeling information should be communicated in OTC labeling.

REFERENCES

1. http://www.fda.gov/AboutFDACentersOfficesOfficeofMedicalProductsandTobacco/CDER/ucm106378.htm
2. http://www.accessdata.fda.gov/scripts/cder/drugsatfda/

ACKNOWLEDGEMENTS

Bayer HealthCare LLC for supporting the research; Rutgers University for funding the research.

Disclosure: Author(s) of this presentation have the following to disclose concerning possible financial or personal relationships with commercial entities that may have a direct or indirect interest in the subject matter of this presentation: Kim Le: Post-doctoral fellow at Bayer HealthCare Consumer Care Meena Ramachandra: Post-doctoral fellow at Bayer HealthCare Consumer Care Lindsay Brust: Nothing to disclose Vrunda Patel: Employee – Bayer HealthCare Consumer Care Michael Toscani: Nothing to disclose